T2375

Stressed Stability Studies of Granulated Metronidazole, Tetracycline HCl, Famotidine and Colloidal Bismuth Subcitrate

Yunqi Wu1 and Reza Fassihi2. 1 SCOLR Pharma, Inc., 2Temple University School of Pharmacy

SCOLR Pharma, Inc.
3625 132nd Ave SE, Suite 300
Bellevue, WA 98006
www.scolr.com

Abstract

To evaluate the stabilities of combined granulation of metronidazole and tetracycline HCl, and famotidine and colloidal bismuth subcitrate (CBS) under stressed conditions. Granules of metronidazole and tetracycline HCl (1:1, w/w), and famotidine and CBS (1:20, w/w) were made using wet granulation method by adding 3% and 1% sodium carboxymethylcellulose (Na-CMC), respectively. Dried granules were stored under stressed storage conditions (40 C, 75%RH). Sampling at predetermined time intervals, metronidazole and tetracycline HCl granules were dissolved in 0.1N HCl, diluted with water, and analyzed by HPLC using a C18 column eluted with methanol-acetonitrile-50 mM KH2PO4, pH 2.5 (10:15:75, v/v/v) at a flow rate of 1.0 ml/min. Famotidine and CBS granules were dissolved in HCl buffer (USP, pH 2.0) and filtered. The solution was diluted with water and analyzed by the same HPLC method. Residuals were dissolved in 70% HNO3 and diluted with water. Colored solution was obtained from 2 ml of CBS solution reacted with 1 ml of 5% thiourea, this sample was diluted with water and detected at 318 nm by UV. Under stressed storage conditions, both metronidazole and CBS showed stable properties in solid state over a period of 90 days. However, significant degradation was occurred in samples containing famotidine with slight degradation of tetracycline HCl. The enhanced temperature associated humidity effect was responsible for the instabilities of tetracycline HCl and famotidine. The degradation rate constant for metronidazole, tetracycline HCl, famotidine, and CBS are 0.0003, 0.0020, 0.0061, and 0.0005 per day, and percent amount remaining after 90 days are 97.76 1.59%, 81.27 4.50%, 57.46 3.43%, and 94.65 3.05%, respectively. Metronidazole and CBS are relatively stable while tetracycline HCl and famotidine are moisture sensitive and should be protected in sealed environment. There was no evidence of incompatibility between the drugs in granular forms in solid state.

Introduction

Metronidazole and tetracycline HCl are two antibiotics used along with bismuth in a triple therapy that is believed to be one of the most effective regimens in the eradication of *Helicobacter pylori* and is used as a "gold standard therapy" (Balaban and Peura, 1997). However, poor patient compliance associated with large number of pills (16 tablets to be taken per day) and many side effects significantly reduces the healing rate, (Malferteiner, 1996; Unge, 1996; Howden, 1997). To overcome these deficiencies, a three layered gastroretentive delivery system based on the gold standard triple therapy with a supplement of an H2 receptor antagonist has been developed to provide for simultaneous delivery of the four actives at different release rates to potentially improve the therapeutic outcome and enhance patient compliance by overcoming the aforementioned limitations.

The physicochemical characteristics as well as the pharmacological and pharmacokinetic parameters of drugs, i.e. metronidazole, tetracycline HCl, and famotidine are well documented.. In addition, owing to the commercial availability of these dosage forms as single doses, they appear to be relatively safe and can be given as combination product. However, one major concern in the proposed delivery system from preformulation point of view is the possible incompatibilities of drugs when combined in a single dosage. Understanding of the nature of actives in combination formulation is the essential component of a successful product development. Drug substances can undergo the decomposition processes via hydrolysis, oxidation, photolysis, etc. When combinations of drugs are used in one formulation, chemical incompatibility issues become one of the most important factors influencing drug stability. In view of the stability of the pharmaceutical substances, adequate stability study is necessary and a requirement prior to submission to the Food and Drug Administration (FDA). In addition, the International Conference on Harmonization (ICH) guidelines provide various techniques that are widely used to monitor the possible decomposition pathways and degradation products. Accelerated/stressed stability tests are often performed to investigate the instability issue of pharmaceutical substances under environment with elevated temperature and humidity in order to facilitate potential chemical reactions and reduce the time period for evaluation of the stability. In this study, potential drug-drug incompatibility is performed using both HPLC and spectroscopic methods.

Methods and Materials

Instruments and Materials:

An HP-1050 series HPLC (Hewlett-Packard, DE) with a UV detector and a SupelcosilTM LC-18 column (4.0 mm 150 mm, 5 m, Supelco, USA), and an HP 8453 diode array spectrophotometer (Hewlett-Packard, DE) were used to perform analytical work. An Accumet 25 pH meter (Fisher Scientific, USA) was used to test pH of solutions. An oscillating sieve machine (Erweka, AR400, Germany), an air oven (Thelco, GCA, USA) were applied in the granulation. Sterifil 47 mm filter system and Millex AH filter membrane were also used in the study (Millipore, Bedford, MA).

Metronidazole (Farchemia, Italy), tetracycline HCl (Sichuan Pharmaceutical Co., Ltd., China), famotidine (Spectrum, NJ) and colloidal bismuth subcitrate (MCP, CT) were the active agents. Sodium carboxy methylcellulose (Na-CMC, Amend, NJ) in low viscosity (31 cps of 2% water solution) was used as binder in wet granulation.

Methanol (MeOH, Spectrum, USA), monobasic potassium phosphate (KH2PO4, Spectrum, USA), phosphorous acid (H3PO4, J. K. Backer, NJ) and sodium hydroxide (NaOH, Amend, NJ) were used to prepare the mobile phase. Nitric acid (70%, ACS, J. K. Baker, NJ), and thiourea (Thiocarbamide, ACS, Sigma, MO) were used to react with bismuth salt in order to produce a colored complex. All the solvent and chemicals were analytical agents and deionized water was used in the study.

Granulation and Storage condition:

Drugs were separated into two sets according to the appearance in different release layers of the tablet and solid-state stability study of the four actives was conducted on granules.
Test was performed over 3 months under the accelerated condition (40 C, 75%RH). Granules of metronidazole and tetracycline HCl were made using wet granulation method by adding 3% (w/v) of sodium carboxy methylcellulose into the blend of powders of metronidazole and tetracycline HCl (1:1, w/w). Granules were dried at a temperature of 60°C and finally sieved with an oscillating granulator.

Same procedures were applied to the preparation of famotidine and colloidal bismuth subcitrate (1:20, w/w) with the binder concentration sodium carboxy methylcellulose at 1% (w/v). Dried granules were sieved, divided into small quantities and accurately weighed (100 mg for granules of metronidazole and tetracycline HCl; 105 mg for granules of colloidal bismuth subcitrate and famotidine).

The accelerated or stressed conditions for stability test were performed. Samples were placed in a capped glass container which contained saturated sodium chloride solution to create a 75% relative humidity (RH) environment (Nyqvist, 1983), and then stored in a constant temperature water bath at 40°C.

Sample Analysis:

Samples (triplicate) were removed at predetermined time intervals. Metronidazole and tetracycline HCl granules were dissolved in 10 ml of 0.1N HCl and instantly diluted to 500 ml of water. Sample solutions were filtered through a 0.45 m filter, and analyzed by the developed HPLC method. Mobile phase was methanol-acetonitrile-50 mM KH2PO4, pH 2.5 (10:15:75, v/v/v) at flow rate of 1.0 ml/min. Analytes were determined simultaneously at 280 nm.

Famotidine and colloidal bismuth subcitrate granules were dissolved in 50ml of HCl buffer (USP, pH 2.0) and filtered with Sterifil 47 mm filter system. Filtrate was diluted with water volumetrically to 500 ml. The solution was analyzed for determination of famotidine by the same HPLC method. Residuals on the filter membrane were collected, dissolved in 2 ml of 70% nitric acid and diluted with water to 100 ml. Accurately measured 2 ml of this solution reacted with 1 ml of 5% thiourea, diluted to 10 ml with water and detected at 318 nm.

Results and Discussion

The stability profiles of metronidazole, tetracycline HCl, famotidine, and colloidal bismuth subcitrate are presented in Figures 1 and 2, respectively.

Figure 1. Stability studies of metronidazole and tetracycline HCl in solid state under accelerated conditions (40°C, 75%RH) over 90 days (n=3).

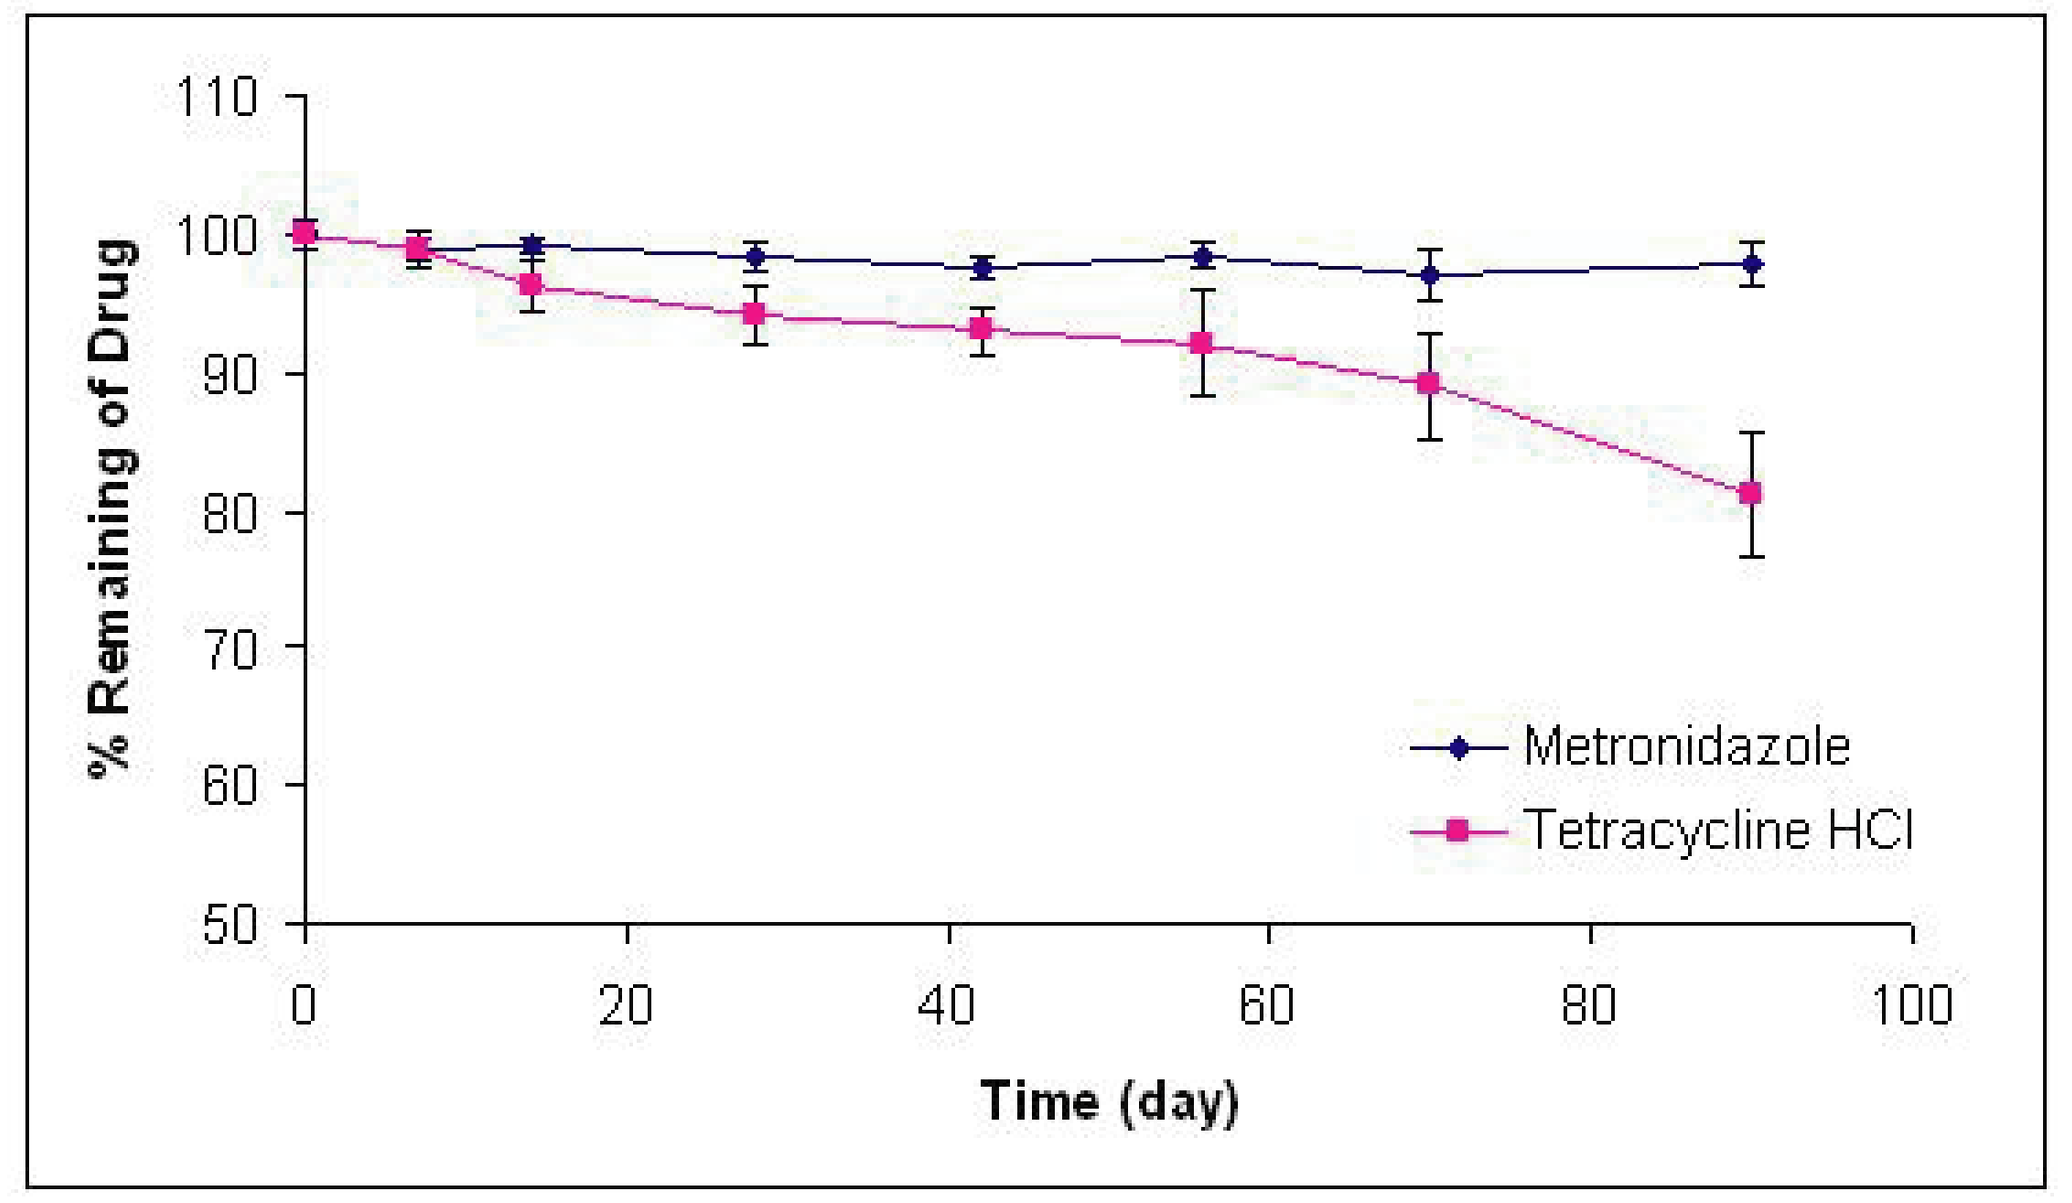
110
100
90
80
70
60
50
% Remaining of Drug
0
20
40
60
80
100
Time (day)
Metronidazole
Tetracycline HCl

Figure 2. Stability studies of famotidine and colloidal bismuth subcitrate (CBS) in solid state under stress conditions (40°C, 75%RH) over 90 days (n=3).

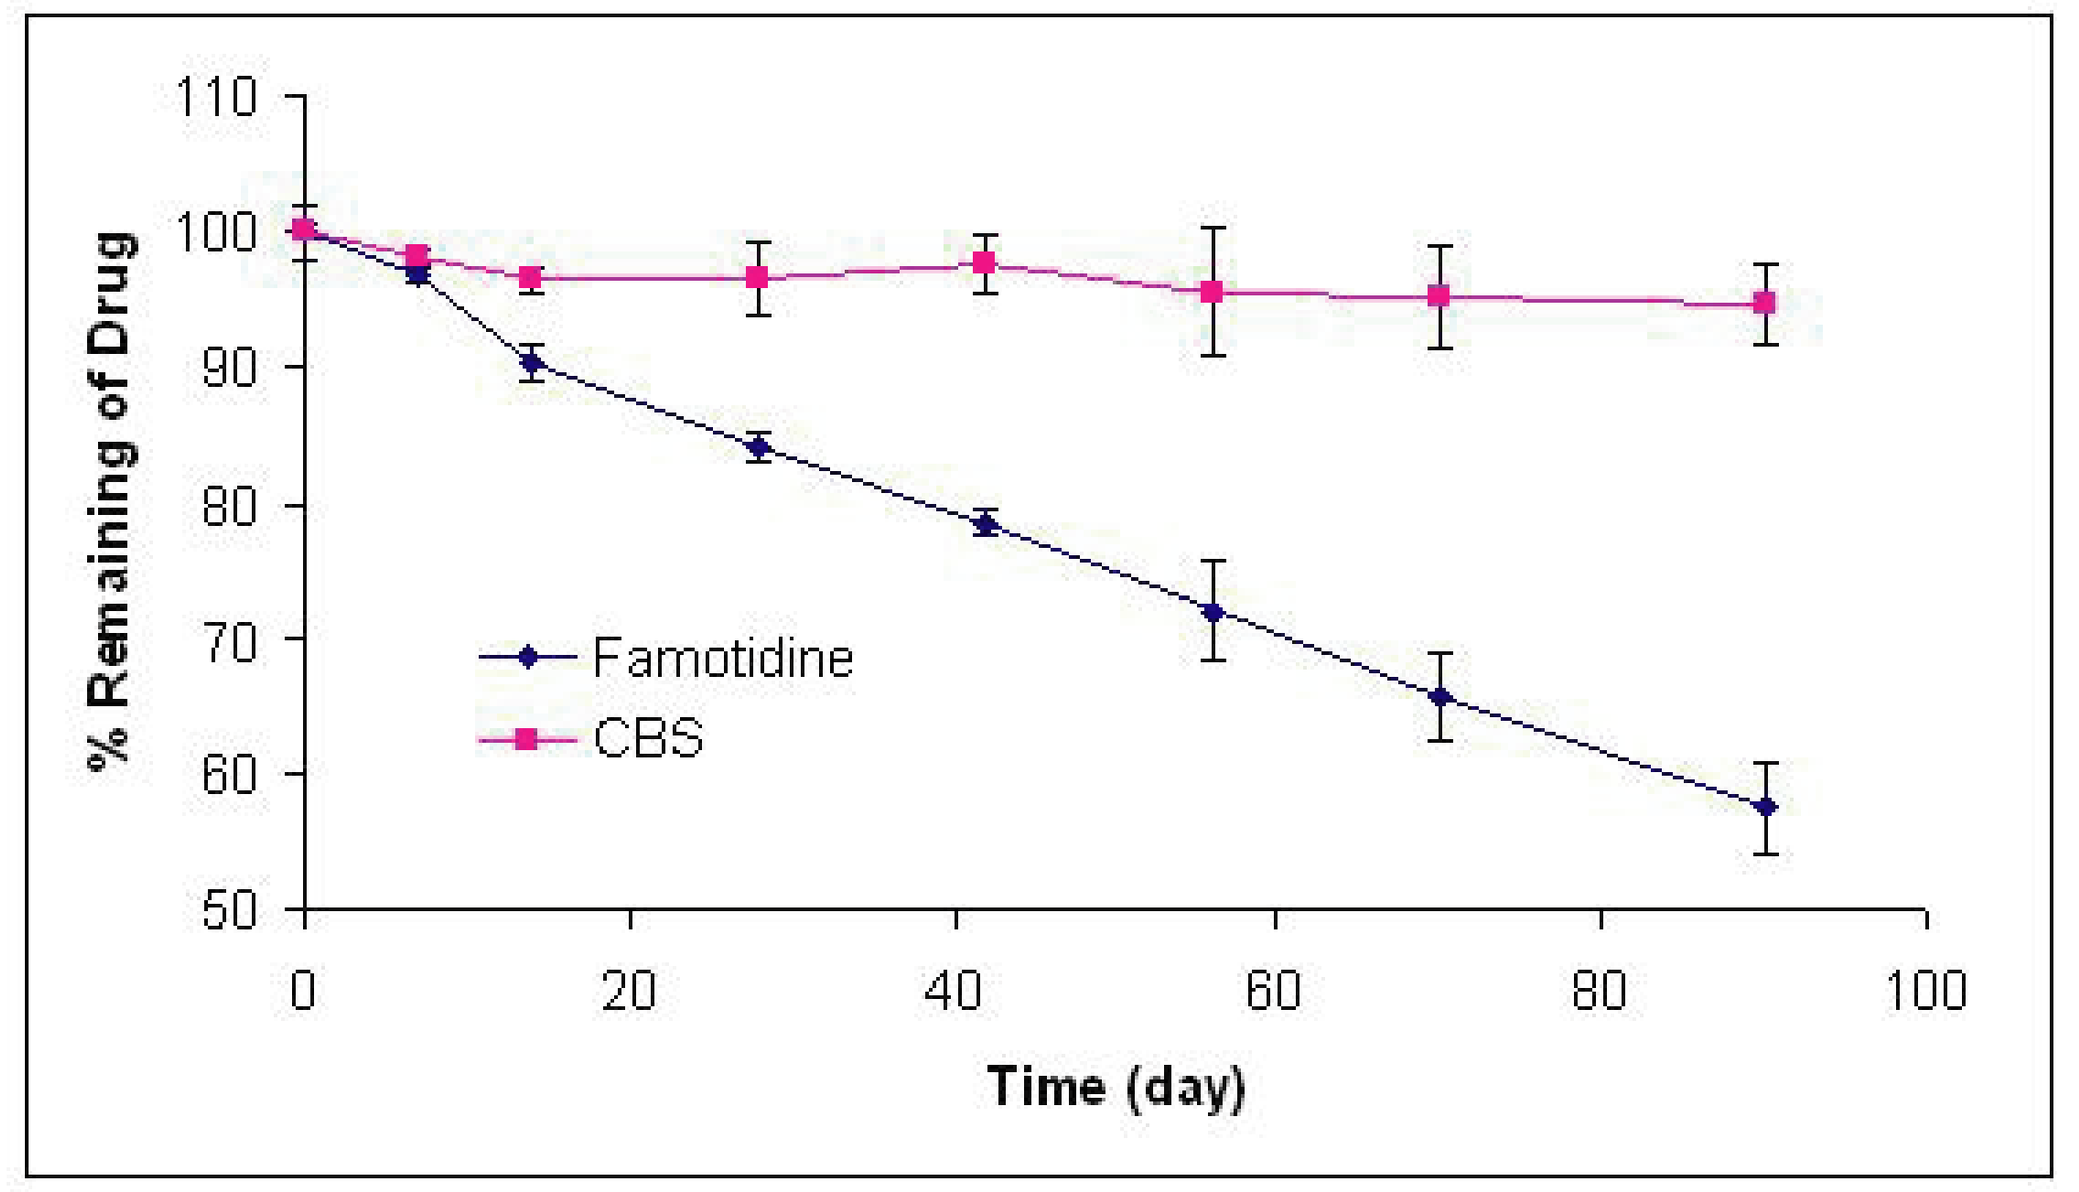
110
100
90
80
70
60
50
% Remaining of Drug
0
20
40
60
80
100
Time (day)
Famotidine
CBS

From the results it appears that metronidazole and colloidal bismuth subcitrate displayed good stabilities in solid state under the accelerated conditions (40 C, 75% RH) over 3 months. Tetracycline HCl, however, was sufficiently stable under normal storage conditions according to previous stability study (Poster presented at AAPS Annual Meeting and Exposition 2003), but under accelerated study underwent slow degradation over the 3 months period of studies. Results further indicated that high temperature and humidity both faciliate tetracycline HCl degradation.

The granules containing tetracycline HCl gradually changed color from light yellow to brown and dark brown after exposure to the humidity and heat over 20-30 days. Results also indicated that light, which is the well-known factor responsible for color transformation of tetracycline HCl, is not the only factor to cause color changes.

Under the given accelerated conditions, a relatively rapid degradation was also observed in the samples containing famotidine. The rate of degradation profile for famotidine demonstrated that famotidine was the least stable drug among the tested active ingredients when subjected to accelerated storage conditions.

Both tetracycline HCl and famotidine underwent degradation in the presence of humidity and elevated temperature. By analyzing the kinetics of degradation, both drugs appear to follow the pseudo-first-order degradation. Nevertheless, metronidazole and colloidal bismuth subcitrate, as shown in Figures 1 and 2, demonstrated negligible degradation in accordance with pseudo-first-order kinetics with very slow degradation rate constant (see Table 1).

Table 1. Degradation Rate Constant, % Remaining after 7 Days and Half-Life for Drugs in Solid State under Stress Condition

Sample	k (day^{-1})	% Remaining after 7 days	$t_{1/2}$ (day)
Metronidazole	0.0003	98.98 ± 0.76	2310
Tetracycline HCl	0.0020	98.85 ± 1.33	346
Famotidine	0.0061	96.92 ± 0.61	114
CBS	0.0005	98.04 ± 0.74	1386

Conclusions

The results of the stability studies in solid state indicated that metronidazole and CBS in the dry state were stable. However, the enhanced temperature associated humidity effect was responsible for the instabilities of tetracycline HCl and famotidine to different extents. It is also evident that under dry conditions there is no potential incompatibility between the drugs in granular forms.
In general in solid state and well-protected storage conditions combinations of metronidazole, tetracycline HCl, famotidine along with colloidal bismuth subcitrate does not present any major problem in the development of a new multi-drug delivery system.

Acknowledgements

Authors wish to thank Dr. Munir A. Hussain from Bristol-Myers Squibb for his input in this work.