W4121 Formulation Development of a Novel Self-Correcting Controlled Release Matrix System Incorporating Film-Forming Polymer Coatings

SCOLR Pharma, Inc.

M. P. Hite[1], S. Turner[1], C. Federici[1], and R. Fassihi[2]
[1]SCOLR, Inc., [2]School of Pharmacy, Temple University

Purpose. To evaluate formulations of a novel self-correcting matrix system displaying bimodal and near zero-order release profiles incorporating coatings of film-forming polymers for the purpose of inducing dissolution lag time and improving manufacturing characteristics. ***Methods.*** Several monolithic formulations were developed using common pharmaceutical excipients such as polyethylene oxide (PEO) and ionizable compounds such as sodium bicarbonate. The formulations were selected for their manufacturability as directly compressible dry blends and for their ability to control hydration rate of the matrix according to the principles of self-correcting ionic gel matrices. Film-forming polymers such as ethyl cellulose and methyl cellulose, and combinations of film forming agents and plasticizers such as polyethylene glycol (PEG), were then applied via fluid-bed apparatus to create coated monolithic dosage forms able to withstand a variety of in-vitro testing conditions. Dissolution studies were conducted using a type II apparatus over 18- and 36-hours. Hardness and friability studies were performed to ensure adequate manufacturability. ***Results.*** The film-forming polymer coatings were well accepted by the delivery system and demonstrated the ability to temporarily suppress dissolution without significantly altering the release profile of the delivery system. The formulations also showed an ability to enhance or suppress dissolution of the system as a whole, and enhance the ruggedness of the delivery system by increasing hardness and decreasing friability of the tablets without adversely affecting the release profile. ***Conclusion.*** Results suggest that the application of film-forming polymer coatings to alter the release profile or to improve the manufacturability of this novel matrix system is possible. Such alterations may be advantageous in suppressing dissolution of soluble actives and improving hardness of controlled release formulations.

Introduction

Film forming coatings and their combinations with hydrophilic matrices are well characterized by the pharmaceutical industry for the purposes of improving the physical characteristics of dosage forms, taste-masking, and the alteration of dissolution profiles, such as the induction of a burst effect, lag time or pH-specific release. Combinations of film forming coatings and self-correcting dosage forms have not been widely investigated, and the purpose of this study was to examine the application of film forming coatings applied to novel monolithic hydrophilic matrix tablets.

Multiple film forming coating formulations were applied to a single manufactured batch of controlled release nicotinic acid tablets employing a self-correcting hydrophilic matrix. A variety of coatings were investigated according to their effect upon dissolution performance: 1) increased hardness and friability while having no effect upon dissolution performance, 2) accelerated dissolution producing pronounced or slight burst effect, 3) temporarily suppressed dissolution resulting in substantially zero-order release thereafter, and 4) suppressed dissolution over extended periods resulting in near-zero order release, and 5) delayed dissolution resulting in up curving release profiles. The amount of tablet weight increase varied from 0.9% to 8.9% according to the purpose of the formulation. Coating formulations were evaluated using a USP Type II dissolution apparatus in 900mL of both acidic media (0.1N hydrochloric acid) and de-ionized water, and hydrodynamic influences were evaluated at 50 and 100 rpm.

Purpose

I. The present study concerns the evaluation of film forming coatings on the dissolution performance of monolithic self-correcting controlled release hydrophilic matrix tablets of nicotinic acid.
II. The present study also evaluates the effect of film forming coatings on the physical characteristics of self-correcting matrix tablets.

Methodology

Materials
Nicotinic acid (niacin) was supplied by Zetapharm, Inc. (New York, NY). Polyethlene Oxide (PEO), Methocel E5 Cellulose Ether (viscosity 4-6 cP*), Methocel E15LV Cellulose Ether (viscosity 12-18 cP*), and Ethocel FP 20 Cellulose Ether was supplied by Dow Chemical (Midland, MI). Sodium Bicarbonate was purchased from Natrium Products (Cortland, NY). Stearic acid was purchased from Ashland Chemical (Santa Anna, CA). Sepifilm™ LP 010 and Sepifilm™ LP 770 were supplied by Seppic Inc. (Fairfield, NJ). *cP (centipoise) is equivalent to milipascal-seconds (mPa-s)

Tablet Preparation
Self-correcting hydrophilic matrix tablets containing 500mg nicotinic acid, 250mg polyethylene oxide, 125mg sodium bicarbonate and 9mg stearic acid were produced at a contract manufacturer (CRM) using a high speed rotary tablet press in standard IPT B-standard caplet-shaped tooling with approximate dimensions of 0.312 in. x 0.750 in. Average uncoated tablet weight was 884 mg.

Coating Preparation
Wurster coating was performed using a Fluid Bed Feasibility Processor (S/N 10218, Fluid-Air, Inc., Aurora, IL) coupled with a Watson-Marlow 101 U/R peristaltic pump (S/N 010.4202.000, Watson-Marlow Bredel Inc., Wilmington, MA) and a Jun-Air 2000-40P air compressor (S/N 450707, Pacific Air Technology, Mission Viejo, CA.

Coating conditions: All coating formulations except Ethocel FP 20-E15LV 2% were applied using 0.5L bowl with Wurster coating apparatus with an average of 30 tablets. Ethocel FP 20-E15LV 2% was applied using 2L bowl with Wurster coating apparatus with an average of 120 tablets. Table 1 contains the specific parameters for each coating formulation.

Table 1.

Coating formulations: Five film-forming coatings of differing compositions were investigated for their effect upon tablet hardness and friability and dissolution performance. Solid dispersions of all coating solutions were mixed under agitation. Methocel E5-PEG and Methocel E15LV-PEG were formed as a 12% solid dispersion (10% Methocel; 2% PEG) in dH2O. Sepifilm™ LP 010 and Sepifilm™ LP 770 were also formed as 12% dispersions (as made available from the manufacturer) in dH2O. Ethocel FP 20-E15LV was formed as a 15% solids dispersion (11.25% Ethocel FP 20; 3.75% Methocel E15LV) in 6.5:3.5 acetone:ethanol solvent mixture.

Hardness and Friability Evaluation
The tablet hardness was performed on a Vankel VK200 hardness tester (S/N 8-1175-1100, Vankel, Cary, NC) using standard testing protocol. The hardness (kP) of tablets was measured and the mean hardness was calculated and reported. The tablet friability was determined on a Vankel single drum friabilator (S/N 4-1931-1100, Vankel, Cary, NC).

Dissolution Studies
Dissolution was performed on a USP Type II dissolution apparatus (Hansen SR-8 Plus, S/N 1100-2057; Erweka DT-70, S/N 10642076C; Vankel 7000, S/N 1-6813-0903, S/N 1-6812-0903, and S/N 1-5737-0301) using 900mL media of either de-ionized water or 0.1N hydrochloric acid at 50 and 100 rpm at 37 degrees C. Spectrophotometric analysis in 0.1N HCl performed as per the Niacin USP 24 monograph method. Absorbance was detected on a UV/VIS spectrophotometer at 275nm (Beckman DU-640, S/N 411969) in 0.1N HCl and 285nm (Cary 50, S/N EL03097618) in dH2O.

Duration of dissolution assay was 18-36 hours, with data points collected every 10-20 minutes during the first two hours and every hour thereafter. After completion of assay, an infinity determination was run at 250 rpm for 1 hour to ensure complete release. Absorbance was corrected for fraction release (%) utilizing the infinity point as the maximum. Dissolution curves were generated utilizing standard methods.

Results and Discussion

This study was conducted in order to evaluate the effect of film forming coatings on self- correcting hydrophilic nicotinic acid tablet matrices. Dissolution studies were conducted for coating formulations C1-8, the results of which are shown in Figures 1-8. Evaluation of physical characteristics (tablet hardness, friability and thickness) were also conducted, the results of which are shown in Table 2.

Among the methylcellulose-based coatings, two film-forming coatings of differing viscosities, Methocel E5 (C1) and Methocel E15LV (C2) containing polyethylene glycol (PEG) as a plasticizer, were investigated for their effect upon tablet hardness and friability, and were shown to have nominal effect upon dissolution performance (Figures 1 and 2).

Figure 1 Dissolution profile of Methocel E5-PEG coated tablets (C1) and uncoated control tablet in 900mL dH2O, 50rpm, 37 degrees C.

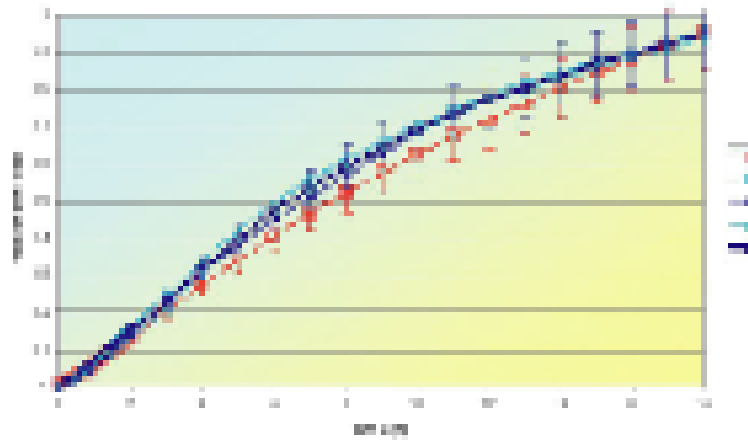

Figure 2 Dissolution profile of Methocel E15LV-PEG coated tablets (C2) and uncoated control tablet in 900mL dH2O, 50rpm, 37 degrees C.

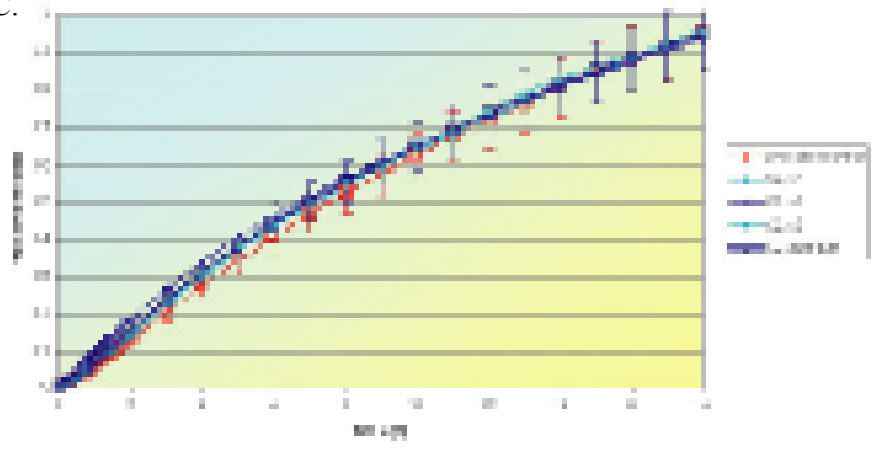

Two additional commercially-available pre-mixed methylcellulose-based coatings containing stearic acid as a plasticizer, Sepifilm™ 010 and Sepifilm™ 770, were also investigated for their effect upon dissolution performance. Sepifilm™ 010 (C3) was shown to provide a slight burst effect during the initial 2 hours and substantially near-zero order release thereafter (Figure 3). Sepifilm™ 770 (C4) was shown to provide a significant burst effect during the first 8-10 hours of release followed by a release profile similar to the control formulation (Figure 4).

Figure 3 Dissolution profile of Sepifilm™ LP 010 coated tablets (C3) and uncoated control tablet in 900mL dH2O, 50rpm, 37 degrees C.

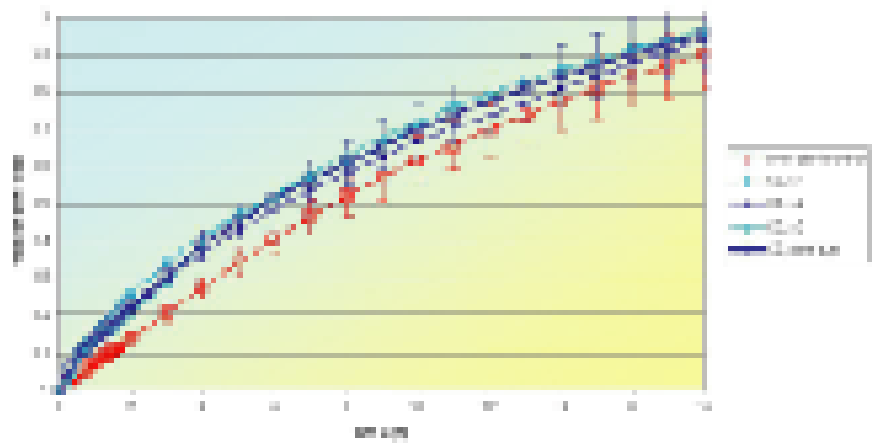

Figure 4 Dissolution profile of Sepifilm™ LP 770 coated tablets (C4) and uncoated control tablet in 900mL dH2O, 50rpm, 37 degrees C.

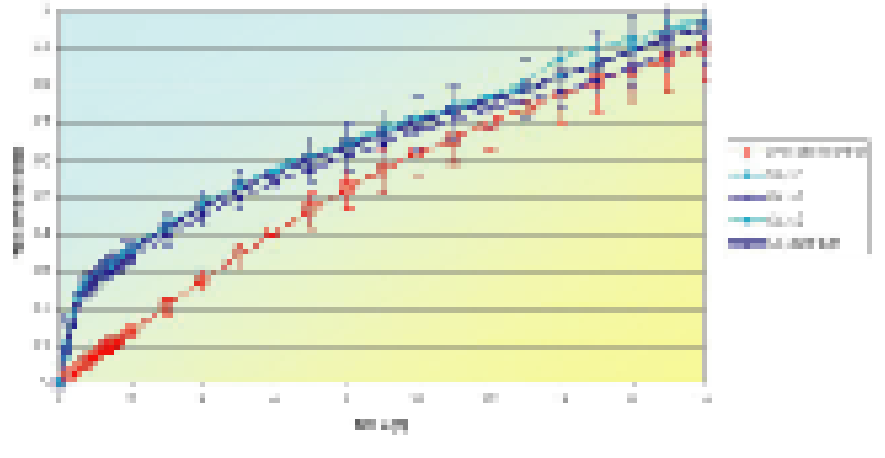

Among the ethylcellulose-based coatings, the effect upon dissolution varied widely with the increase in tablet weight. Using Ethocel FP 20/E15LV at 0.9% tablet weight increase (C5), a temporary suppression of dissolution is induced during the initial 3 hours, and thereafter the release profile is identical to the uncoated reference formulation (Figures 5a and 5b).

Figure 5a Dissolution profile of Ethocel FP 20/E15LV coated tablets (C5) at 0.9% tablet weight increase and uncoated control tablet over 0-18 hours in 900mL 0.1N HCl, 50rpm, 37 degrees C.

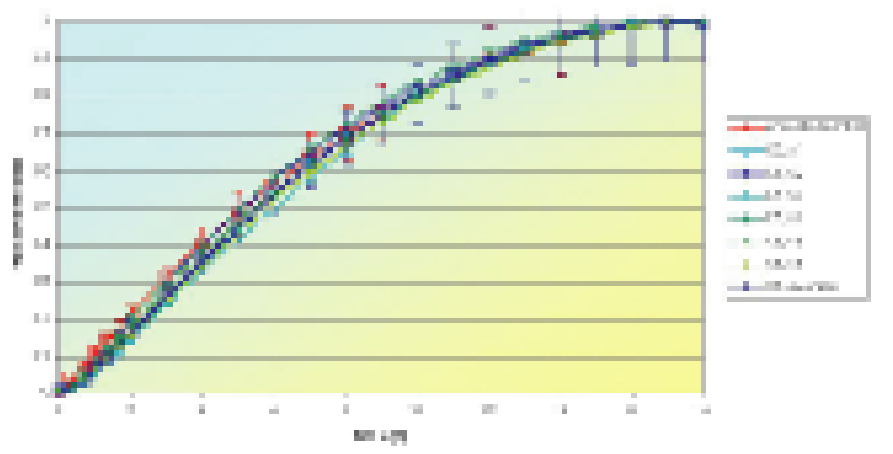

Figure 5b Dissolution profile of Ethocel FP 20/E15LV coated tablets (C5) at 0.9% tablet weight increase and uncoated control tablet over 0-3 hours in 900mL 0.1N HCl, 50rpm, 37 degrees C.

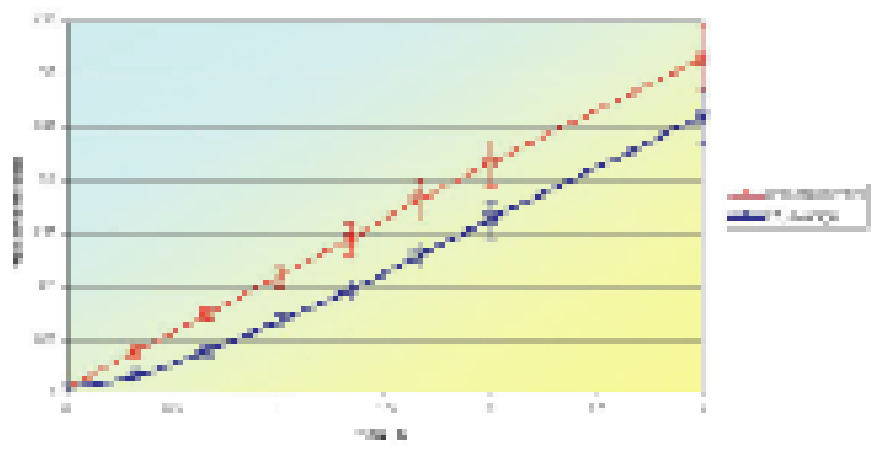

Using Ethocel FP 20/E15LV at 2% tablet weight increase (C6), dissolution is suppressed and the release kinetics of the dissolution profile are extended to near-zero order over 36 hours, compared with an 18-hour profile observed in the reference formulation (Figure 6).

Figure 6 Dissolution profile of Ethocel FP 20/E15LV coated tablets (C6) at 2.0% tablet weight increase and uncoated control tablet over 36 hours in 900mL dH2O, 50rpm, 37 degrees C.

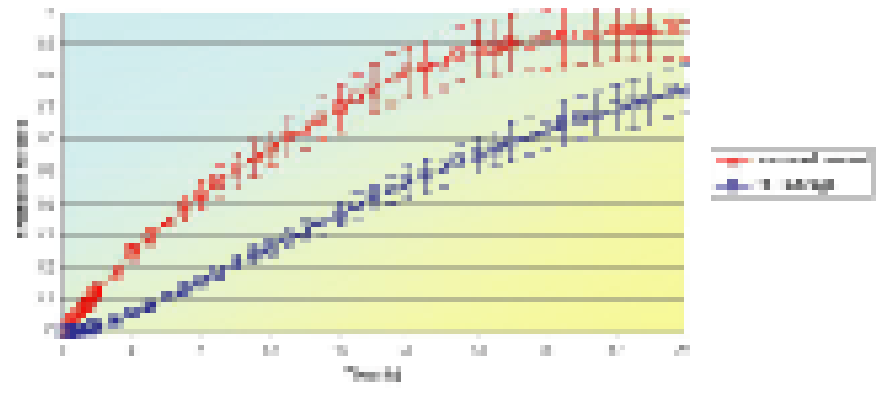

Using Ethocel FP 20/E15LV at much higher tablet weight increases of 5.8% (C7) and 6.8% (C8), the dissolution profile is delayed such that less than 30mg of nicotinic acid (<6% of drug load) is released during the first 8 hours, and thereafter a distinct up curving release profile is observed, (Figure 7). The effect of hydrodynamics and media variation upon these higher tablet weight increase coatings was also evaluated: acidic media produced significantly more variation at 5.8% than at 6.8% tablet weight increase; increased agitation of 100rpm in acidic media produced similar effects at both 5.8% and 6.8% tablet weight increase (Figure 8).

Figure 7 Dissolution profile of Ethocel FP 20/E15LV coated tablets at 5.8% (C7) and 6.8% (C8) tablet weight increase and uncoated control tablet over 18 hours in 900mL 0.1N HCl, 50rpm, 37 degrees C.

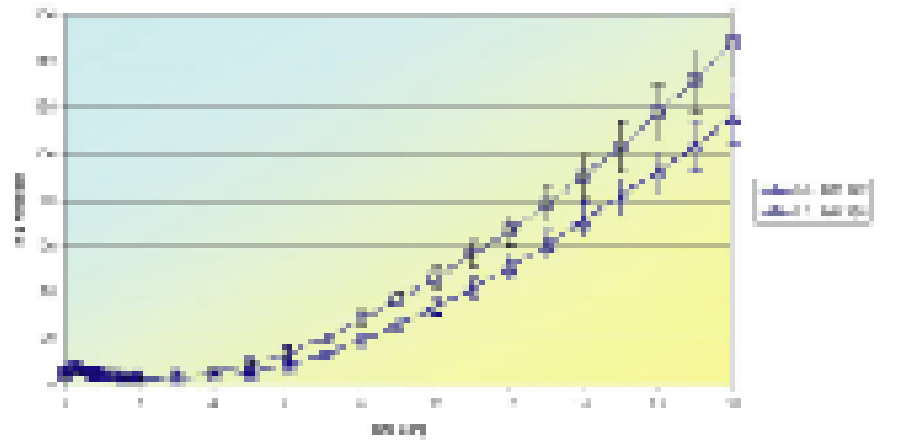

Figure 8 Dissolution profile of Ethocel FP 20/E15LV coated tablets at 5.8% (C7) and 6.8% (C8) tablet weight increase and uncoated control tablet over 36 hours in 900mL 0.1N HCl, 50rpm and 100rpm, 37 degrees C.

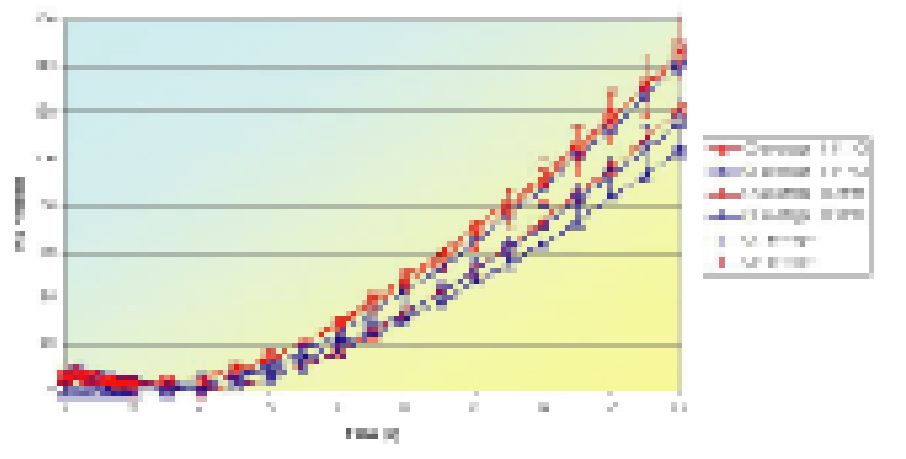

Physical evaluation of hydrophilic matrix tablets possessing film forming coatings were also conducted and compared to uncoated reference tablets, (Table 2). The methylcellulose-based coatings containing PEG as a plasticizer (Methocel E5 and E15LV) and the high weight increase ethylcellulose coatings (5.8% and 6.8%) displayed greatly increased hardness values (increases of 10.6-12.0 kp) relative to the uncoated formulations. The non-pigmented methylcellulose-based coating containing stearic acid as a plasticizer (Sepifilm 010) and the lower weight ethylcellulose coatings (2%) displayed smaller increases in hardness (increases of 2.9-4.6 kP). The pigmented methylcellulose-based coating containing stearic acid as a plasticizer (Sepifilm 770) and the lowest weight increase ethylcellulose coatings (0.9%) showed only nominal increase in hardness values. The friability of the uncoated control formulations was excellent, and thus negligible improvements in friability of the coated formulations were observed. Increases in thickness relative to uncoated tablets displayed a trend largely consistent with the increases in tablet weight among the coatings, with tablet weight increases of 8.9%, 6.8%, 5.8% and 4.4% corresponding to thickness increases of 0.027 and 0.023, 0.020, 0.019 and 0.012 in., respectively. Tablet weight increases of 6.3%, 2.0% and 0.9% slightly deviated from this trend, corresponding to thickness increases of 0.017, 0.004 and 0.007 in.; such variation may result from differences in thickness of uncoated tablets or uneven coating at very low coating volumes.

Table 2. Physical Evaluation of Coatings C1-7.

Conclusions

The self-correcting hydrophilic matrix nicotinic acid tablets used in this study were shown to be amenable to film forming coatings. These film forming coatings were shown to be capable of improving tablet hardness, which may potentially improve manufacturability when employed with poorly compressible formulations. Through the use of methylcellulose-based coatings containing PEG as a plasticizer, such improvements in tablet hardness may be achieved with no impacting the dissolution profile of the hydrophilic matrix. These film forming coatings were also shown to be capable of altering the in vitro dissolution profiles of a monolithic tablet formulation, through a temporary suppression of dissolution without affecting the remaining release profile, an induction of a slight or dramatic burst effect, a sustained suppression of dissolution resulting in extended near-zero order release, or a prolonged delay in dissolution resulting in an up curving dissolution profile.

References

In Vivo - In Vitro Correlation (IVIVC) of a Novel Monolithic Controlled Release Dosage Form
S. J. Turner, M. Hite, C. Federici, R. Fassihi. AAPSPharmSci Vol. 4, No. 4, Abstract W4203 (2002)

Development of a High Drug Load Monolithic Controlled Release Oral Delivery System for Niacin: A Novel Approach. S. Turner, M. Hite, C. Federici R. Fassihi. AAPS PharmSci Vol 3, No. 3, Abstract T3248 (2001)

METHOCEL in Aqueous Systems for Tablet Coating. 1995. Dow Chemical. Form 198-00755-0300-X GW

ETHOCEL FP Ethycellulose Polymers product specification sheet. 2000. Dow Chemical. Form 198-02001-700X-AMS

Sepifilm™ LP. 2001. Seppic Inc.

Title	Formulation Development Of A Novel Self-correcting Controlled Release Matrix System Incorporating Film-forming Polymer Coatings View Full Text
Author(s)	Michael Patrick Hite[1], Steven Turner[1], Catherine Federici[1], Reza Fassihi[2] [1] SCOLR, Inc., [2] School of Pharmacy, Temple University
Journal	AAPS PharmSci
	Vol. 5, No. 4, Abstract W4121 (2003)
ISSN	1522-1059
Publisher	American Association of Pharmaceutical Scientists